An Interdisciplinary Approach in a Rural Community Pharmacy Achieves Positive Patient Outcomes While Increasing Billable Patient Care Opportunities

Geoffrey Twigg, PharmD, BCACP, CDE; John Motsko, RPh, CDE; Jeffrey Sherr, RPh, FACA

Apple Discount Drugs & Core Clinical Care, 404 North Fruitland Blvd., Salisbury, MD 21801

INTRODUCTION

Introduction: Diabetes education has been especially challenging in the tri-county area of the lower Eastern Shore of Maryland. This area has been traditionally under served in regards to healthcare. The incidence of diabetes in the area is in excess of 17%, approaching twice the national average[1]. The area is also under served by primary care providers and diabetes specialists. This model positions pharmacists in a position to help alleviate this gap in care.

Objective: To build a financially stable, replicable, and interdisciplinary program around a Center of Excellence (COE) model in a rural, community pharmacy. The implementation of the model would result in an increase in the number of patients seen, improved outcomes for those patients, and an increase in the number of billable clinical services the pharmacy provides.

Methods: A grant from the Community Pharmacy Foundation allowed exploration of possible business models to form a COE. Apple Discount Drugs formed a new corporation based on the Center of Excellence model inside of a community pharmacy. The COE provides comprehensive medication management and disease state education using an interdisciplinary team of pharmacists, a physician, a nurse practitioner, and a registered dietician. The new COE used its ability to offer medication therapy management (MTM) and diabetes self management education and training to market to referring prescribers and third party payers to expand the number of billable, clinical services that the pharmacy offered. Patients were counseled according to the National Standards for Diabetes Education[2] and the core components of MTM[3].

RESULTS

As a result of the new model, the pharmacy has acquired additional billing opportunities. The pharmacy's largest commercial third party payer has granted the pharmacist/CDEs a limited "provider status" to offer DSME/T (diabetes self management education and training). The patient centered medical home (PCMH) associated with that third party payer is now a large referral source into the pharmacy and has given the clinical pharmacists access to the patient's clinical portal containing all documentation relevant to clinical care. Having healthcare professionals on staff that third party payers recognize as providers has increased the ability of the pharmacy to directly bill for more of the clinical services offered. A total of 309 patients were seen at least once during the study period and 120 graduated the Diabetes Center DSME/T 10 hour training course.

A1C LEVEL CHANGES*

A1C Level	Pre-DSME	Post-DSME	Change
High	15.8	10.5	5.3
Average	8.72	7.43	1.29
Low	5.8	5.5	0.3

*99 Patient Records

BMI CHANGES*

BMI	Pre-DSME	Post-DSME	Change
High	54.2	49.7	4.5
Average	35.68	32.4	3.28
Low	19.3	18.1	1.2

*82 Patient Records

PATIENT DEMOGRAPHICS

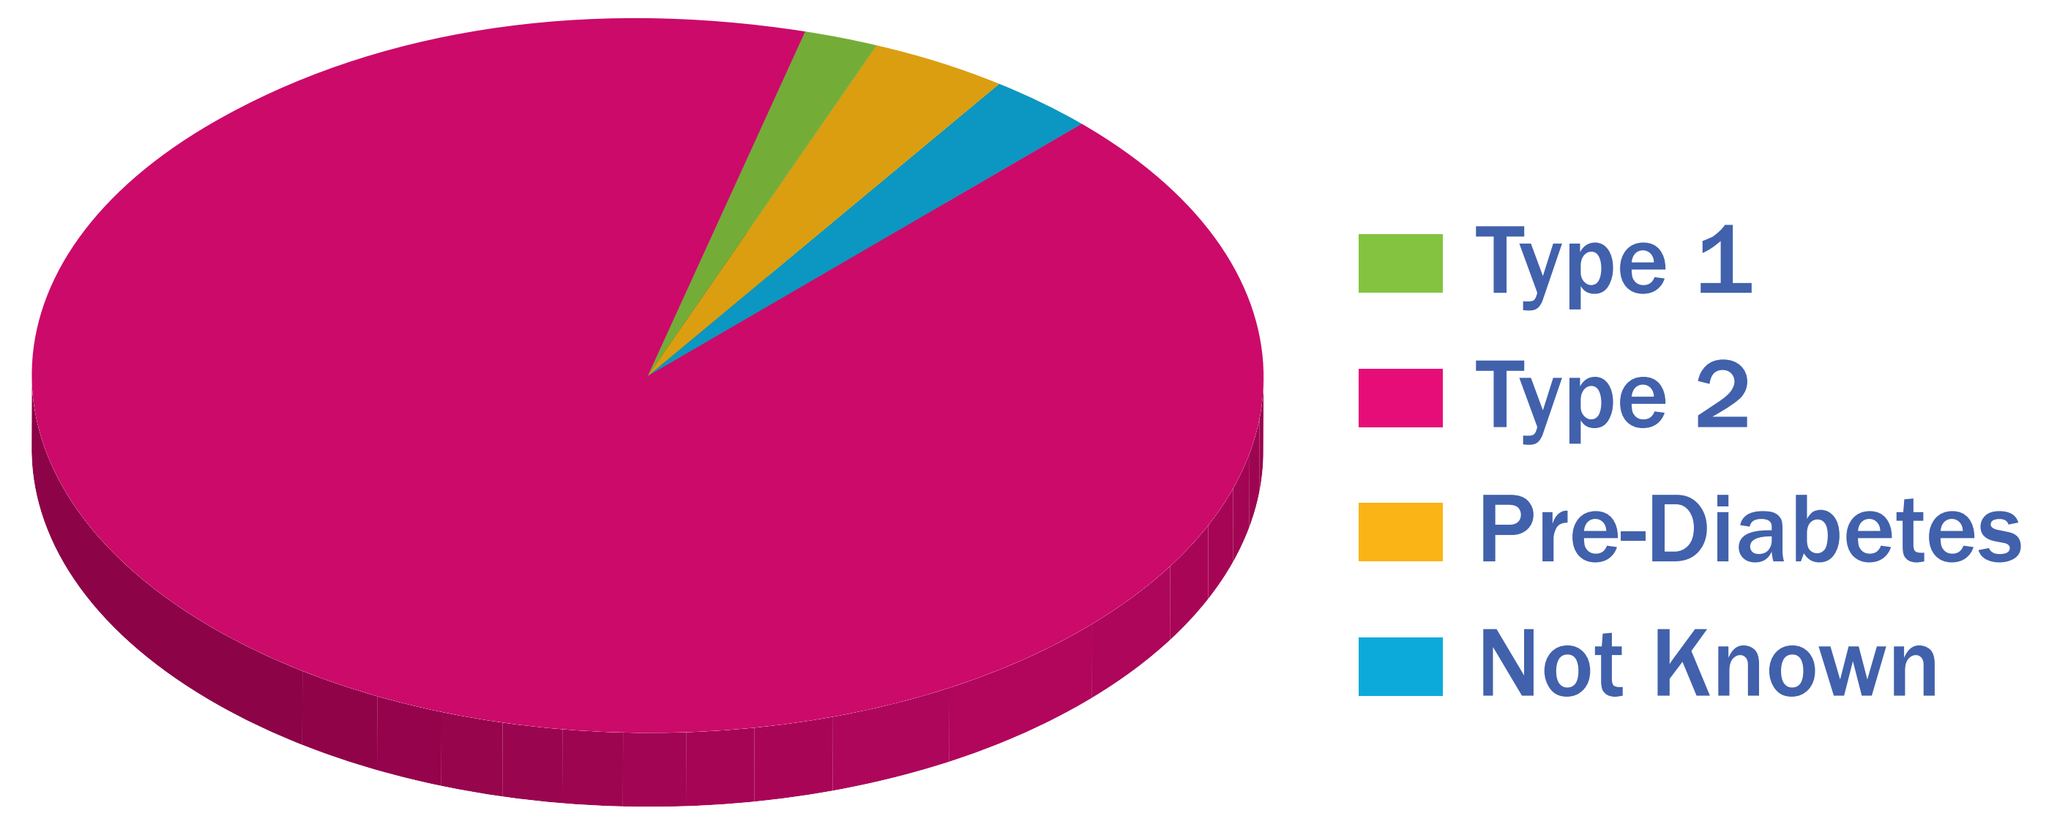
Type 1
Type 2
Pre-Diabetes
Not Known

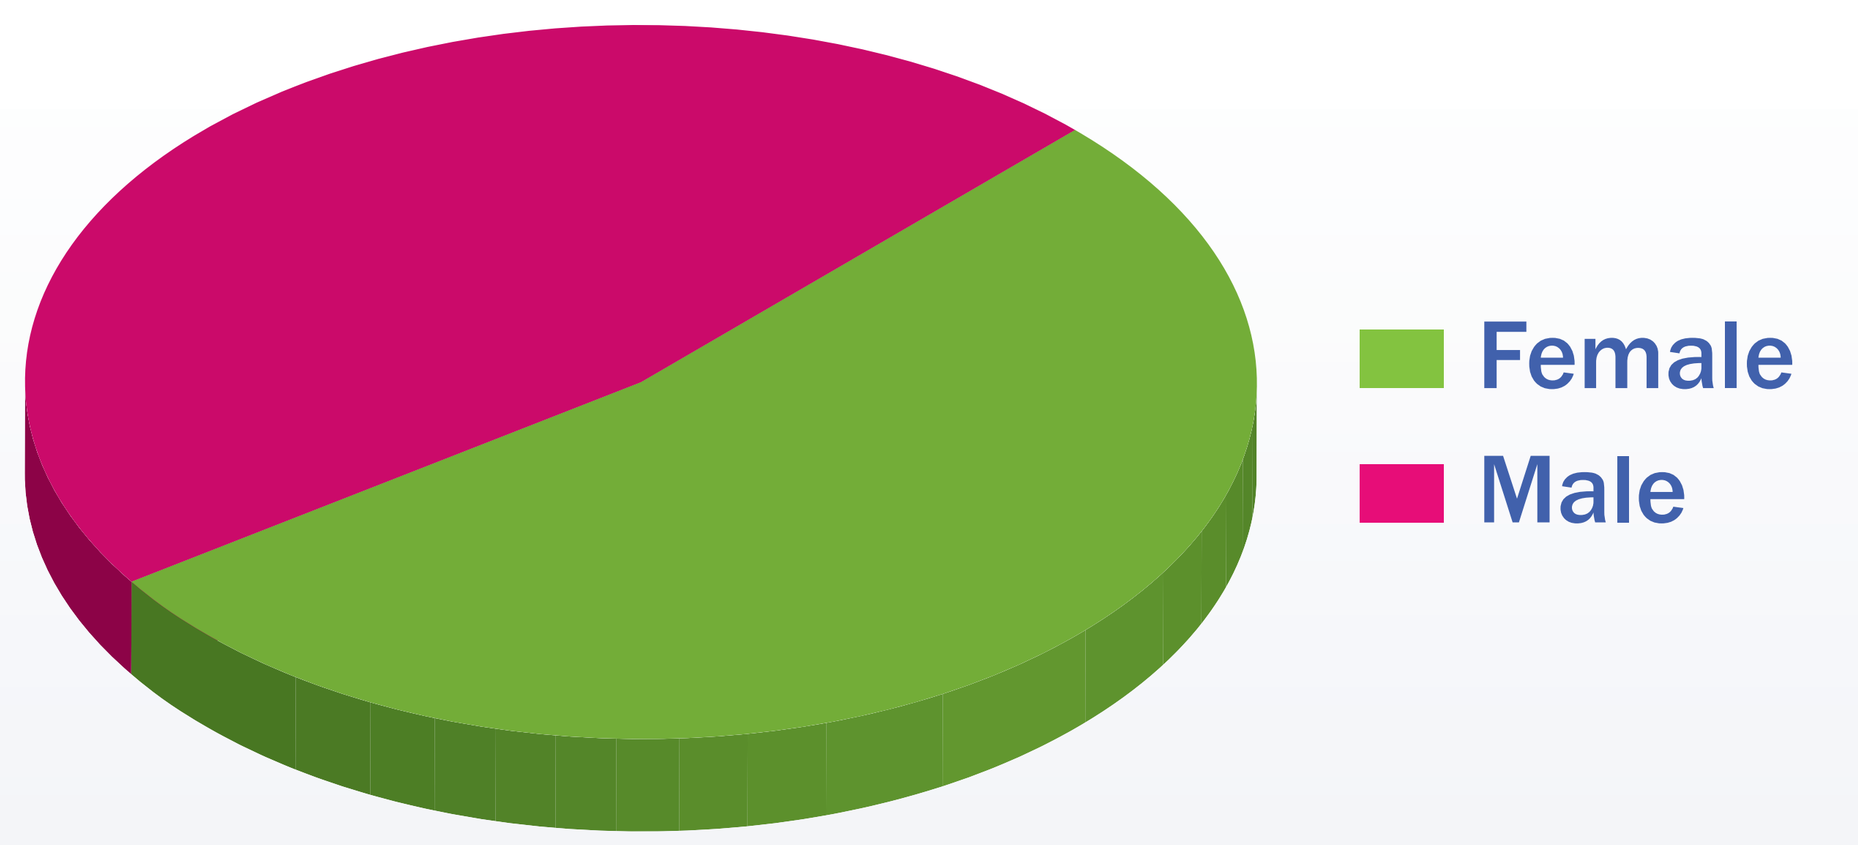
Female
Male

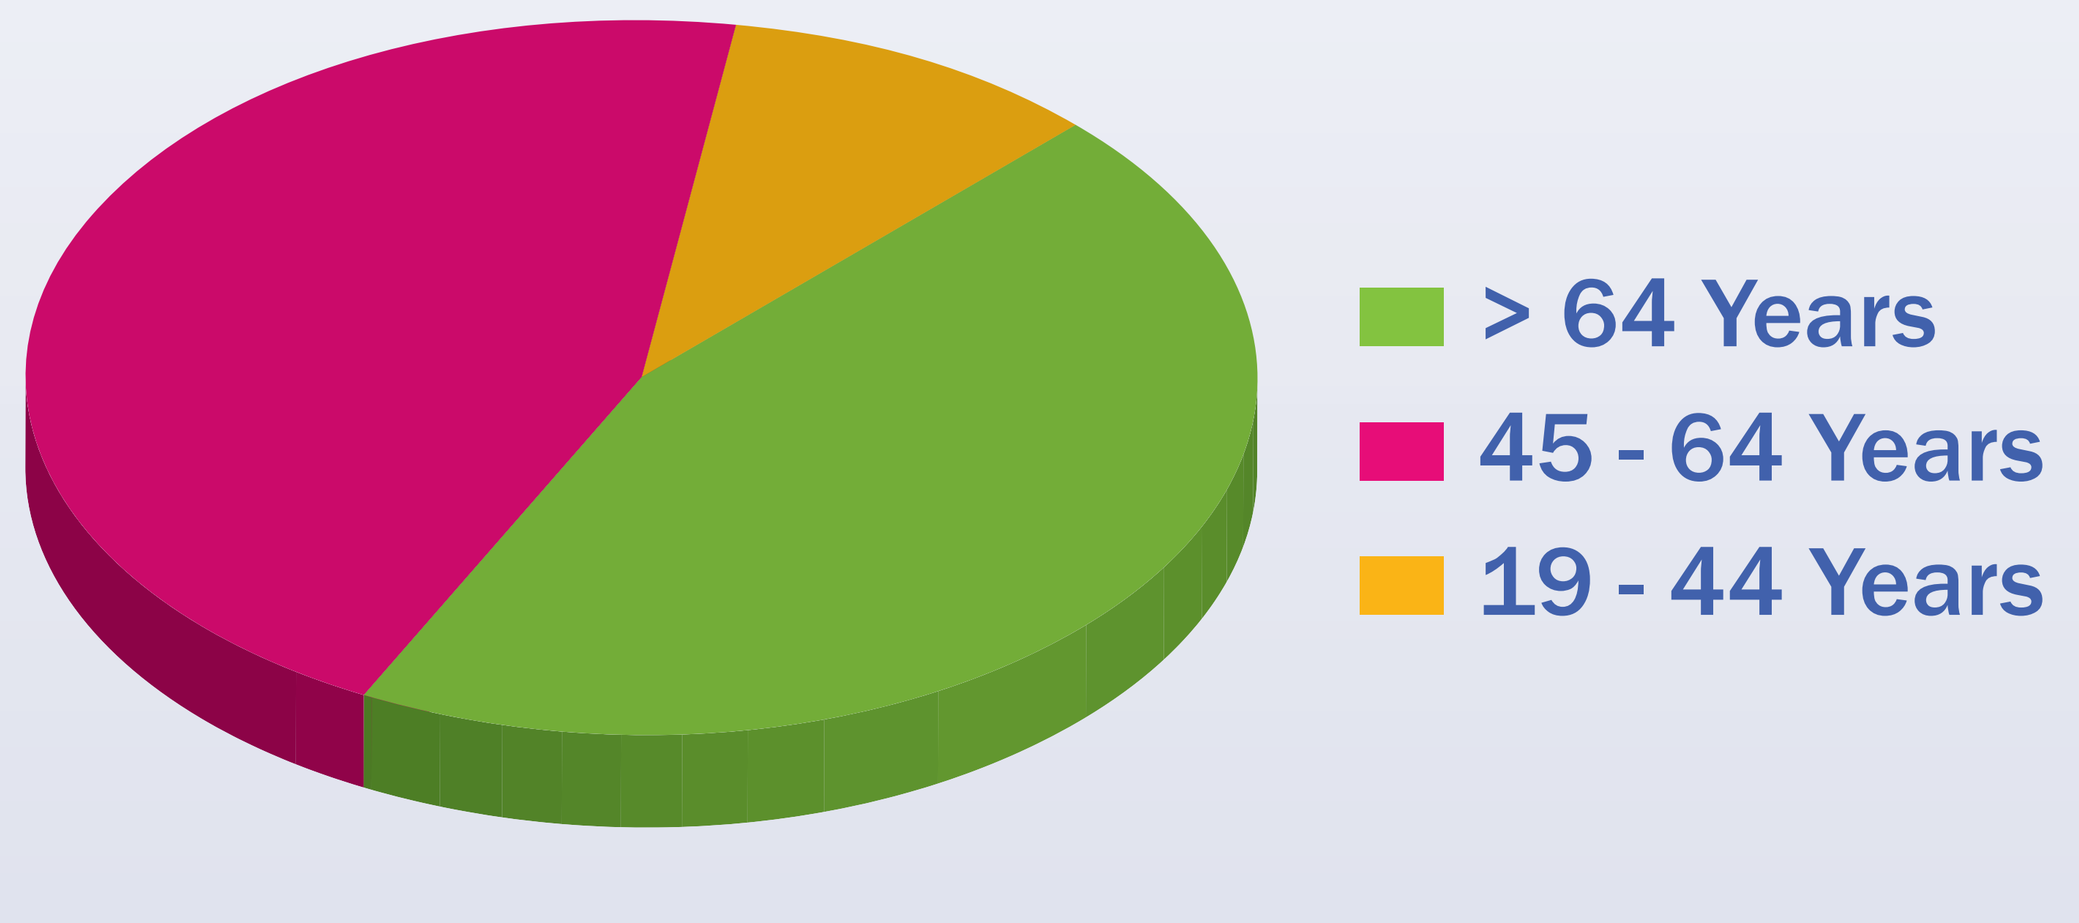
> 64 Years
45 - 64 Years
19 - 44 Years

Discussion

The practice of community pharmacy is continually evolving. With lowering margins for reimbursement in traditional dispensing, pharmacy must identify new areas of financial growth and stability. With pharmacists being one of the most trusted and easily assessable health care professionals, they have a unique ability to affect patient outcomes.

With additional reimbursement opportunities available, the pharmacy has been able to increase both the number of hours devoted to clinical services and expand the number and types of patients seen in its clinical department. This increase has corresponded to positive outcomes for the pharmacy's patients.

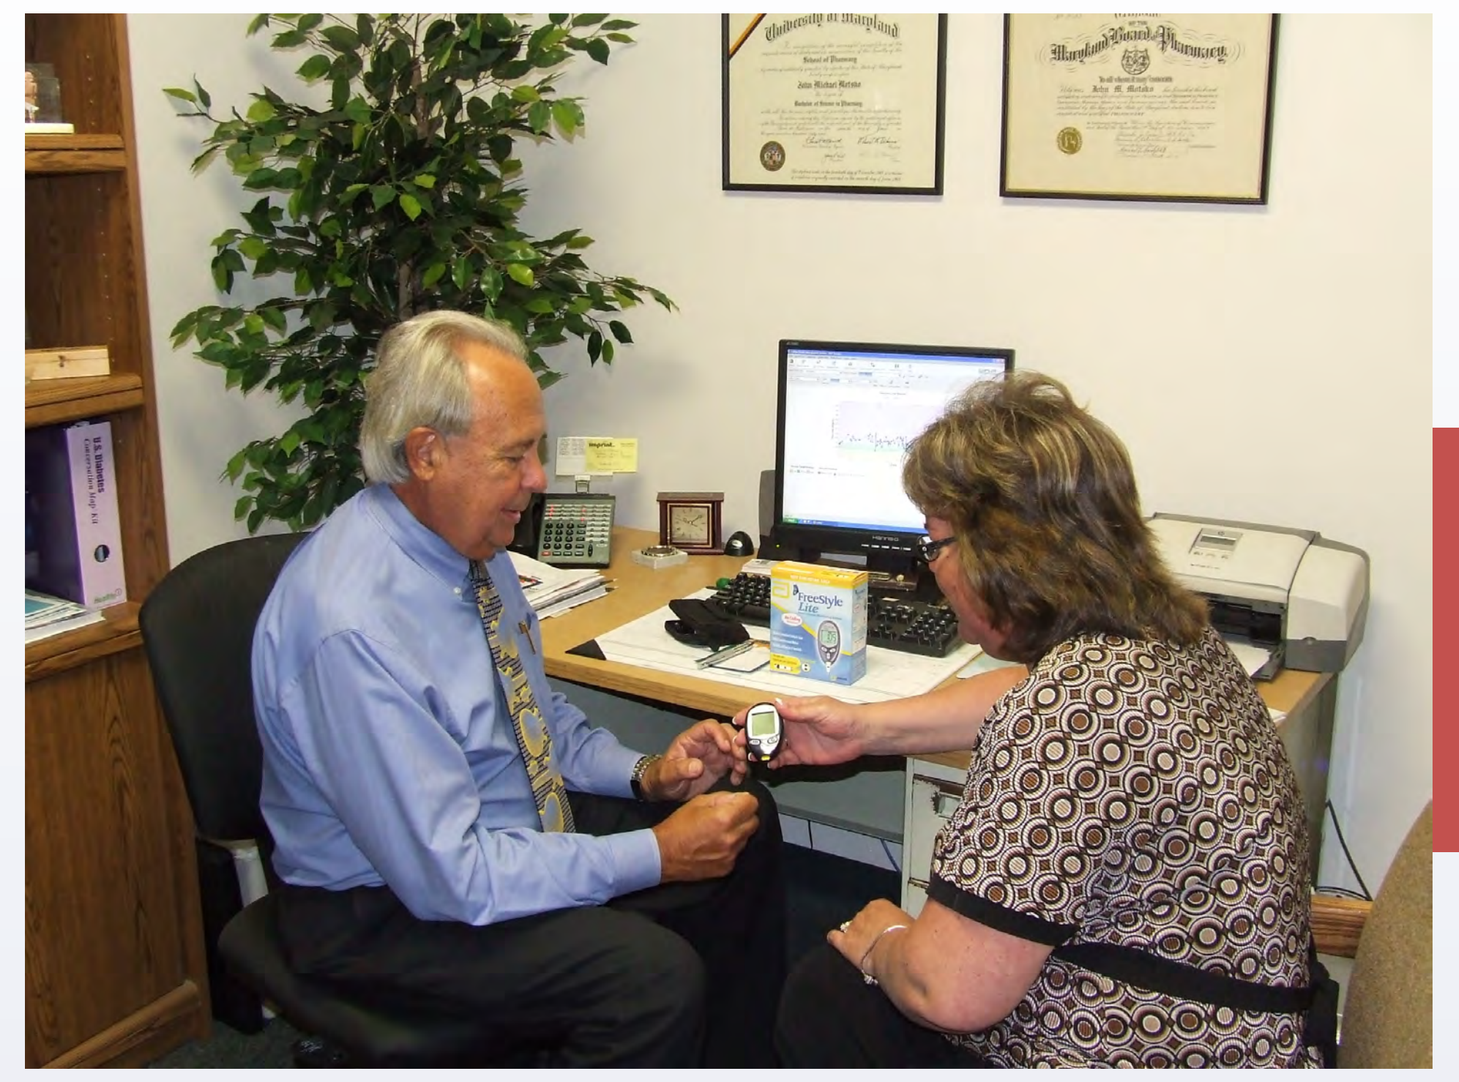

One-on-one consultation

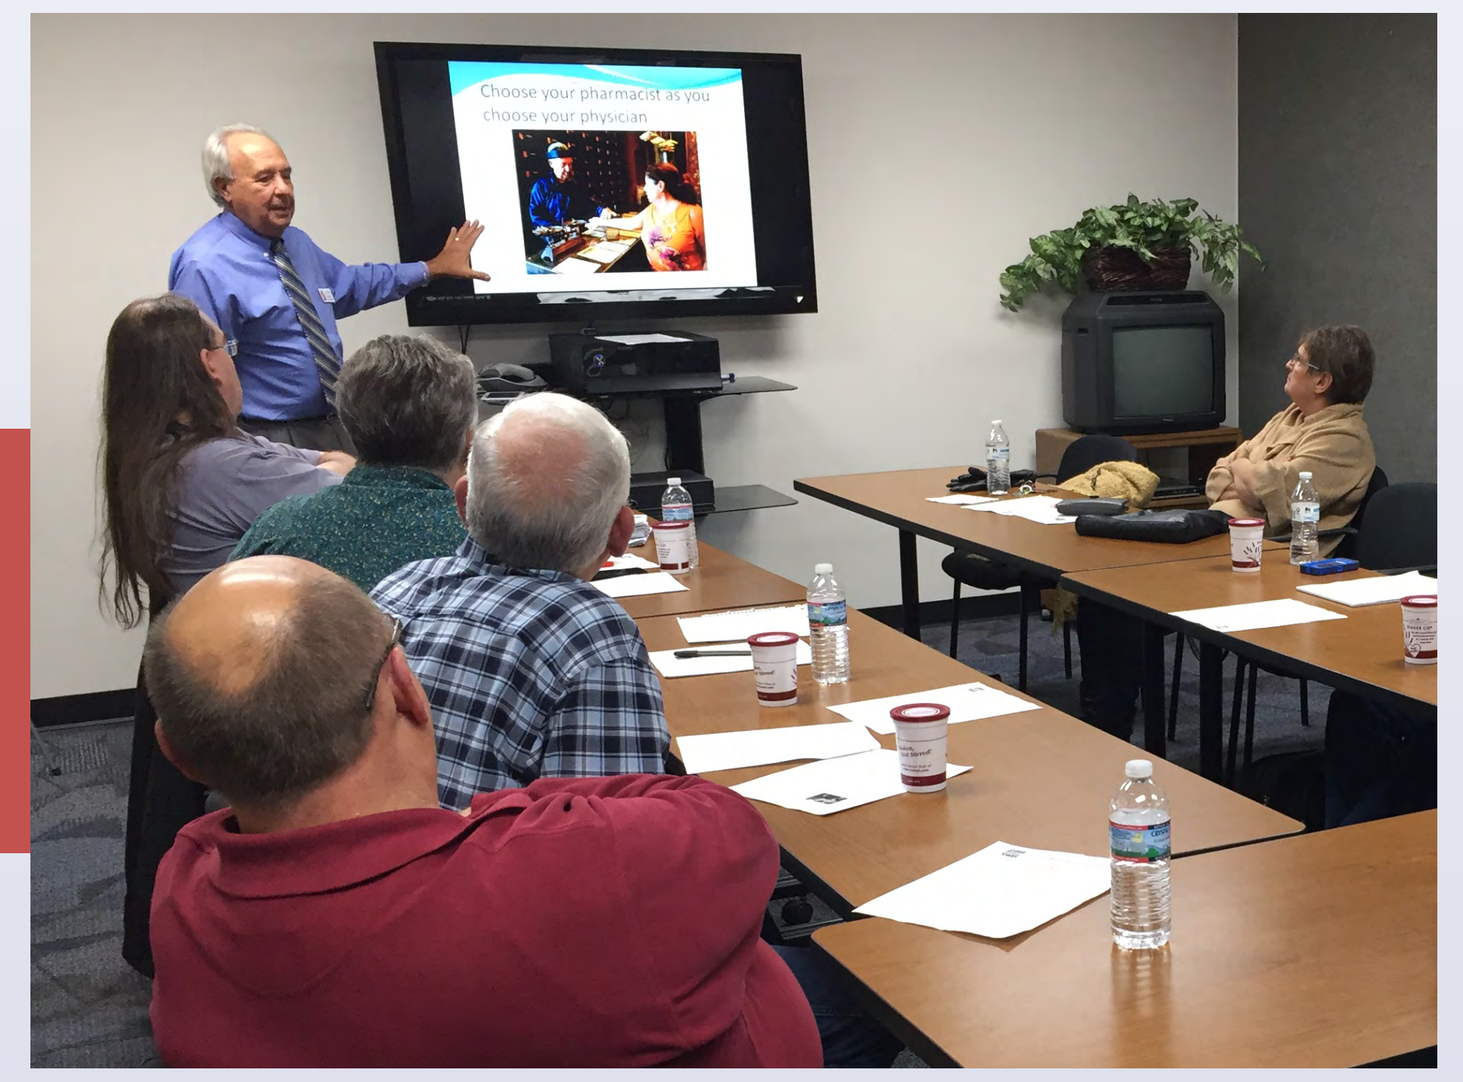

Diabetes training class

Limitations

Positive clinical outcomes have resulted in certain payers recognizing the pharmacy as a provider for certain clinical services with opportunities for growth in others. There are still significant barriers to the pharmacy billing incident to another clinical provider's visit in our rural setting. This is further complicated by commercial payers not having a common claim and credentialing process.

CONCLUSION

Creating an interdisciplinary team business model inside of a community pharmacy can increase not only the number of billable opportunities but also expand the quality of services it can provide and attract new patients and increase referrals from rural providers by improving patient outcomes.

References

[1]Maryland Department of Health and Mental Hygiene. What is the Prevalence of Diabetes? http://phpa.dhmh.maryland.gov/dpcp/SitePages/Prevalence.aspx. Published April 2015. Accessed 13 January 2016.

[2]Haas, Linda, Maryniuk, Melinda, Beck, Joni, et al. National Standards for Diabetes Self Management Education and Support. http://care.diabetesjournals.org/content/37/Supplement_1/S144.full.pdf+html. Diabetes Care. 2014 Jan;37 Suppl 1:S144-53.doi: 10.2337/dc14-S144

[3]Burns, Anne, et al. Medication Therapy Management in Pharmacy Practice: Core elements of an MTM service model (version 2.0). J Am Pharm Assoc. 2008;48:341-353. Doi: 10.1331/JAPhA.2008.08514

Acknowledgements

Apple Discount Drugs and Core Clinical Care would like to thank the Community Pharmacy Foundation for their help and support in undertaking this project.